Dose-Ranging Study of Paraxanthine Ingestion on Cognition, Executive Function, and Psychomotor Vigilance

Dickerson, B.[1], Xing, D.[1], Yoo, C.[1], Jenkins, V.[1], Nottingham, K.[1], Leonard, M.[1], Gonzalez, D.[1], Ko, J.[1], Faries, M.[1,2], Kephart, W.[3], Purpura, M.[4], Jager, R., FISSN[4], Wells, S., FISSN[5], Sowinski, R.[1], Rasmussen C. J.[1], & Kreider, R. B., FISSN[1], [1]Exercise & Sport Nutrition Lab, Texas A&M University, College Station, TX; [2]Texas A&M AgriLife Extension, Texas A&M University; [3]Department of Health Science, University of Wisconsin-Whitewater, Whitewater, WI; [4]Increnovo LLC, Milwaukee, WI; [5]Ingenious Ingredients L.P., Lewisville, TX

Abstract

Background Acute ingestion of 200mg of paraxanthine (PX) affects measures of short-term memory, reasoning, response time to cognitive challenges, and as well as help sustain attention. The optimal and minimal effective dose of PX is currently unknown. The purpose of this study was to assess the efficacy and safety of different doses of PX on markers of cognition, executive function and psychomotor vigilance to one week of continued supplementation.

Methods In a randomized, double blind, placebo-controlled, crossover, and counterbalanced manner, 12 healthy male and female subjects (22.7±4 years, 165±7 cm, 66.5±11 kg, 24.4±3 kg/m^2) were assigned to ingest 200 mg of a placebo (PLA), 50 mg of PX (ENFINITY™, Ingenious Ingredients, L.P.) + 150 mg PLA, 100 mg PX + 100 mg PLA, or 200 mg of PX. Participants completed stimulant sensitivity and side effect questionnaires and donated a fasting blood sample. Participants then performed the Berg- Card Sorting task test (BCST) that is an executive function test that assesses long thought, including reasoning, learning, executive control, and attention shifting; the Go/No-Go test (GNG) that assesses sustained attention and response control through reaction time and accuracy to visual stimuli; the Sternberg task test (STT) that assesses short-term/working memory involving cognitive control processes using reaction time and accuracy; and, the Psychomotor Vigilance Task Test (PVTT) that assesses sustained attention reaction times through responses to visual stimuli. Participants then ingested on capsule of PLA or PX treatments with 8 ounces of water. Participants completed side effects and cognitive function tests after 1, 2, 3, 4, 5, and 6 hours of after ingestion of the supplement. Participants continued ingesting one dose a day for six more days of the assigned supplement and then returned to the lab to donate a fasting blood sample. After a 7-day washout period, participants returned to the lab to repeat the experiment. Participants conducted this protocol two additional times until all four treatments were assessed. IBM SPSS for Windows version 25.0 software (Chicago, IL) was used to analyze data. Data were analyzed by a General Linear Model (GLM) univariate analyses with repeated measures using weight as a covariate and assessing mean and percent changes from baseline with 95% Confidence Intervals (CI's).

Results STT 4-Letter Length Present Reaction Time tended to differ among groups (p-0.06). Assessment of mean changes from baseline with 95% CI's revealed several significant differences among treatments in BCST Correct Responses, Preservative Errors (PEBL), and Preservative Errors (PAR Rules) providing some evidence that PX at varying doses enhanced thought, reasoning, learning, executive control, and attention shifting. There was also evidence of significant differences among treatments in GNG Tasks in Mean Accuracy and response time markers under various conditions assessed providing some evidence that PX influences helps sustain attention. Likewise, there were significant differences among treatments at several timepoints of increasing complexity among STT variables assessed suggesting that PX enhanced the ability to store and retrieve random information from short-term memory of increasing complexity to a greater degree. Finally, there was evidence from the PVTT assessment that response time improved over the series of 20 trials assessed as well as over the course of the 6-hour experiment in the PX treatment suggesting that PX helped sustain attention. Benefit compared to PLA were seen with each dose studied but more consistent effects appeared to be at 100mg and 200mg doses. No significant differences were observed in side effects or standard clinical chemistry panels.

Conclusion Results provide some evidence that acute ingestion of 100 mg and 200 mg of PX may affect some measures of short-term memory, reasoning, response time to cognitive challenges, and as well as help sustain attention and that 7-days of PX ingestion is not associated with any clinically significant side effect.

Background

Many individuals seek ways to improve cognitive function in order to get them through their day. Dietary supplementation with paraxanthine has been purported as one such solution. However, limited data is one of the main concerns of use. This study seeks to examine the efficacy of a paraxanthine supplement on cognition and executive function.

Methods

- Randomized, double-blind, counterbalanced, crossover
- 12 men (n=3) and women (n=9) participated in the study
- Participants met baseline characteristic criteria (22.7±4 years, 165±7 cm, 66.5±11 kg, 24.4±3 kg/m^2)
- Participants supplemented with each of the following for one week followed by a 7-14 day washout period :
 - [PLA] wheat flour (200 mg)
 - [PX+PLA] Paraxanthine (50 mg), Wheat Flour (150 mg)
 - [PX+PLA] Paraxanthine (100 mg), Wheat Flour (100 mg)
 - [PX] Paraxanthine (200 mg)
- Fasting and post-treatment blood samples, pre and post-treatment side effects questionnaires; pre and post treatment cognitive function tests were all collected at Day 1 and Day 7 of each supplemental protocol.

Statistical Analyses

IBM SPSS for Windows version 25.0 software (Chicago, IL) was used to analyze data. Data were analyzed by a General Linear Model (GLM) univariate analyses with repeated measures using weight as a covariate and assessing mean and percent changes from baseline with 95% Confidence Intervals (CI's).

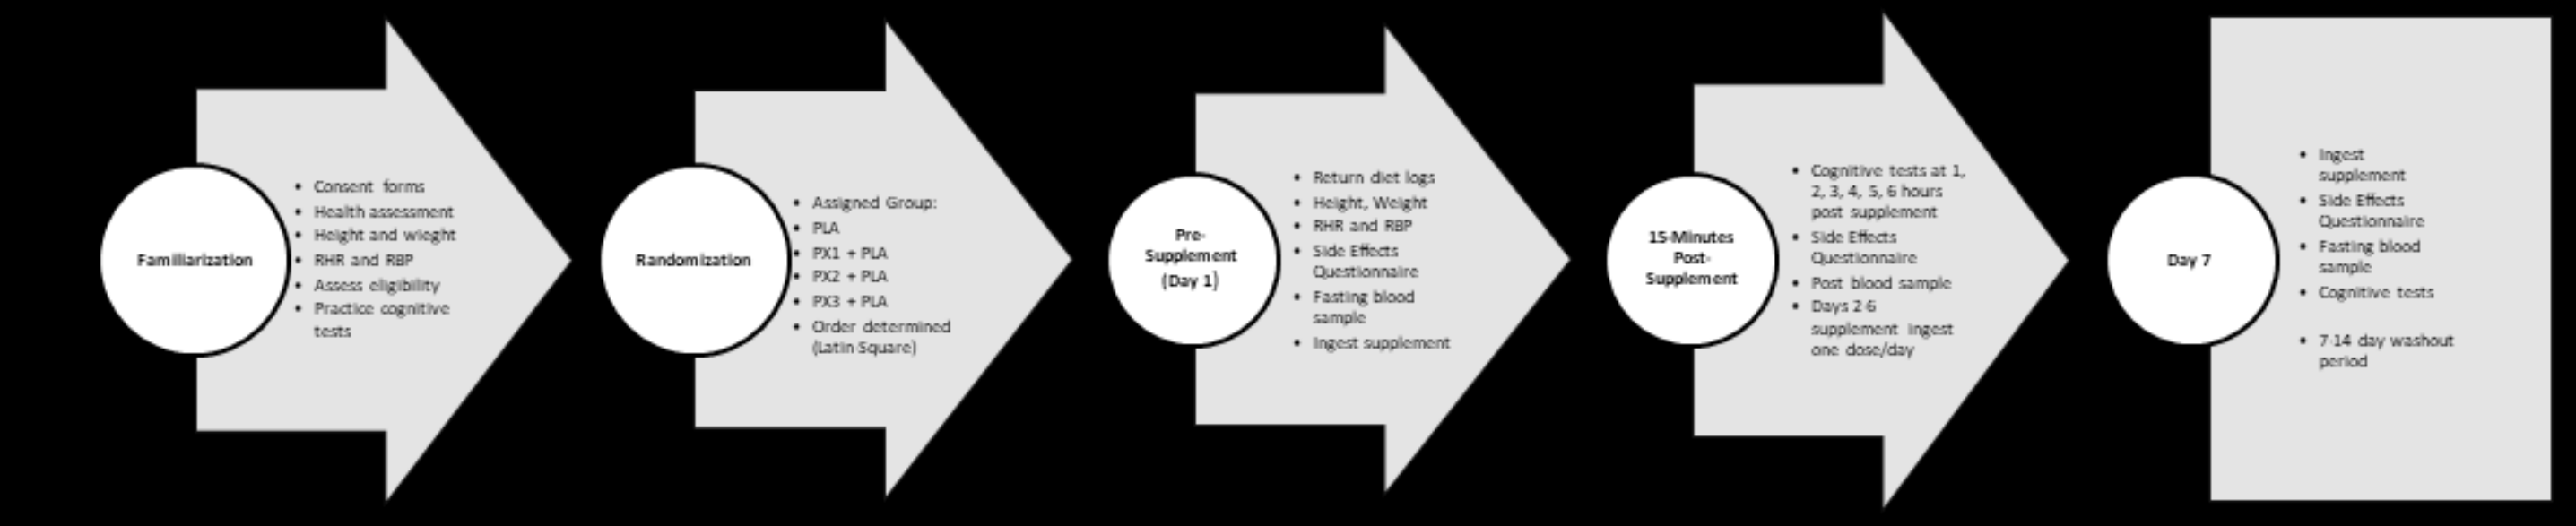
Familiarization
Consent forms
Health assessment
Height and wieght
RHR and RBP
Assess eligibility
Practice cognitive tests
Randomization
Assigned Group:
PLA
PX1 + PLA
PX2 + PLA
PX3 + PLA
Order determined (Latin Square)
Pre-Supplement (Day 1)
Return diet logs
Height, Weight
RHR and RBP
Side Effects Questionnaire
Fasting blood sample
Ingest supplement
15-Minutes Post-Supplement
Cognitive tests at 1, 2, 3, 4, 5, 6 hours post supplement
Side Effects Questionnaire
Post blood sample
Days 2-6 supplement ingest one dose/day
Day 7
Ingest supplement
Side Effects Questionnaire
Fasting blood sample
Cognitive tests
7-14 day washout period

Results

- Several significant differences in BCST, GNG, STT, and PVTT at varying doses with PX supplementation at different timepoints.
 - Significant differences in mean changes from baseline indicated in Preservative Errors (BCST), mean accuracy and response time (GNG), variables of increasing letter length complexity (STT), and reaction time over the course of multiple trials (PVTT) (all p<0.05)
 - Benefits compared to placebo were seen with each dose but most consistent effects seen with 100mg and 200mg doses

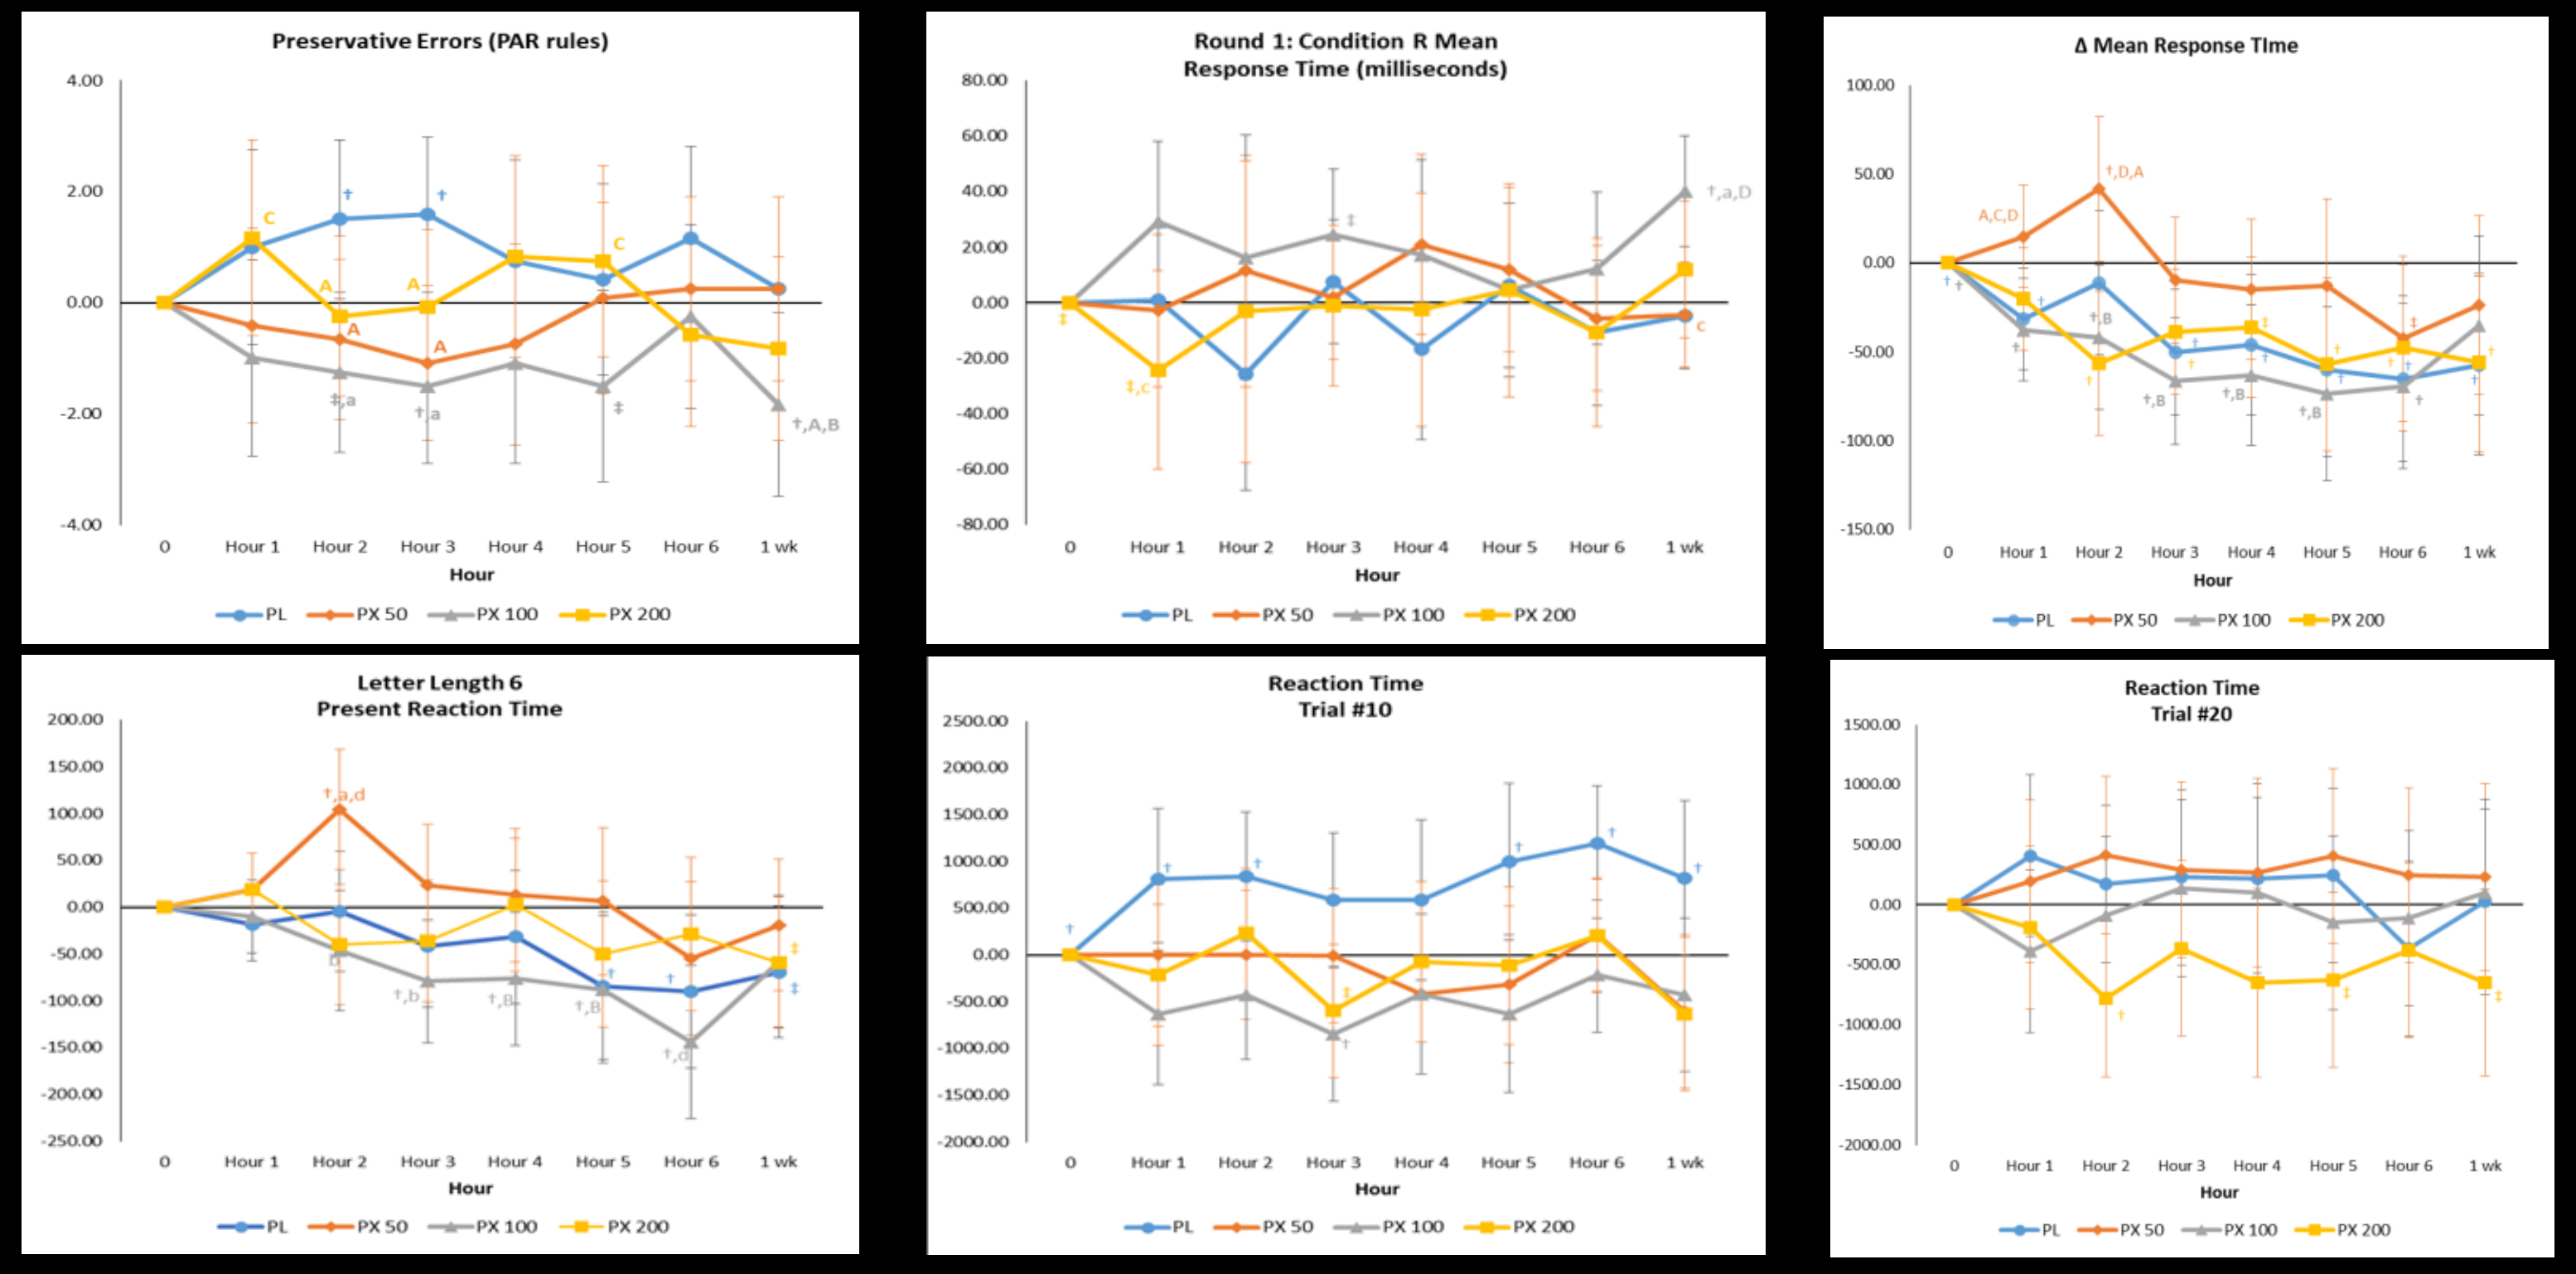
Preservative Errors (PAR rules)
Round 1: Condition R Mean Response Time (milliseconds)
Δ Mean Response Time
Letter Length 6 Present Reaction Time
Reaction Time Trial #10
Reaction Time Trial #20
0
Hour 1
Hour 2
Hour 3
Hour 4
Hour 5
Hour 6
1 wk
Hour
PL
PX 50
PX 100
PX 200

† = p<0.05 difference from baseline value, ‡ = 0.05<p<0.10 difference from baseline, a = difference from placebo, b = difference from PX50, c = difference from PX100, d = difference from PX200; lowercase: p ≤ 0.05, uppercase: 0.05 ≤ p ≤ 0.10

Conclusions & Applications

- Supplementing with PX at various doses provides beneficial effects on cognitive and executive function.
- Significantly beneficial effects were seen at various time points after ingestion and a week after supplementation.
- Most consistent significantly different findings were seen with 100mg and 200mg PX doses.
- Further research is necessary regarding effective dose over a prolonged and fed state.

Acknowledgements & Disclosures

This study was funded as a fee for service project to the Human Clinical Research Facility research by Ingenious Ingredients, LP (ING2). RJ, SDW and MP are co-owners of ING2 and have been named as inventors for the use of PX by ING2.